Optimizing Immune Responses through Targeted Delivery of Neoantigens: A Study on the Dual-Function STRIKE2001 Antibody and Peptide Tag Variants

STRIKE Pharma

Alexandros Kostakis[1,2], Ida Lauren[2], Rosanne Veerman[1], Aman Mebrahtu[3], Johan Rockberg[3], Pierre Dönnes[1,4], Sara Mangsbo[1,2]

[1] Strike Pharma AB, Uppsala, Sweden, [2] Uppsala University, Uppsala, Sweden, [3] KTH Royal Institute of Technology, Stockholm, Sweden, [4] SciCross AB, Skövde, Sweden

UPPSALA UNIVERSITET

Background

Antibodies are excellent carriers of drug cargo to specific cells or organs in the body. Antibody-Drug Conjugates (ADCs) utilize various chemical engineering techniques and linker technologies designed to both deliver and release drugs at specific sites in the body and have historically been used mainly for the delivery of toxic payloads or radionuclides. However, drugs with poor half-life and with specific class-based biodistribution, such as peptide- and oligonucleotide-based therapeutics, could also benefit from targeted delivery. **We have developed a methodology that makes use of biotechnology engineering and high-affinity interactions to build a new version of ADCs, providing the opportunity to adapt the cargo and enable precision medicine-based ADC development.**

Our first drug candidate is STRIKE2001, a humanized IgG2 antibody comprising two peptide-binding single-chain variable fragments (scFvs) linked to the CH3 domain. These scFvs bind to a short non-immunogenic peptide tag (pTag). A peptide of choice is then synthesized by solid-phase peptide synthesis along with the pTag, creating a flexible tumor antigen delivery system coined **the Adaptable Drug Affinity Conjugate (ADAC) platform, which is an excellent alternative to the immunogenic biotin-avidin approach used in many studies.** We have also studied the biological activity of the tag itself, and the data are presented herein.

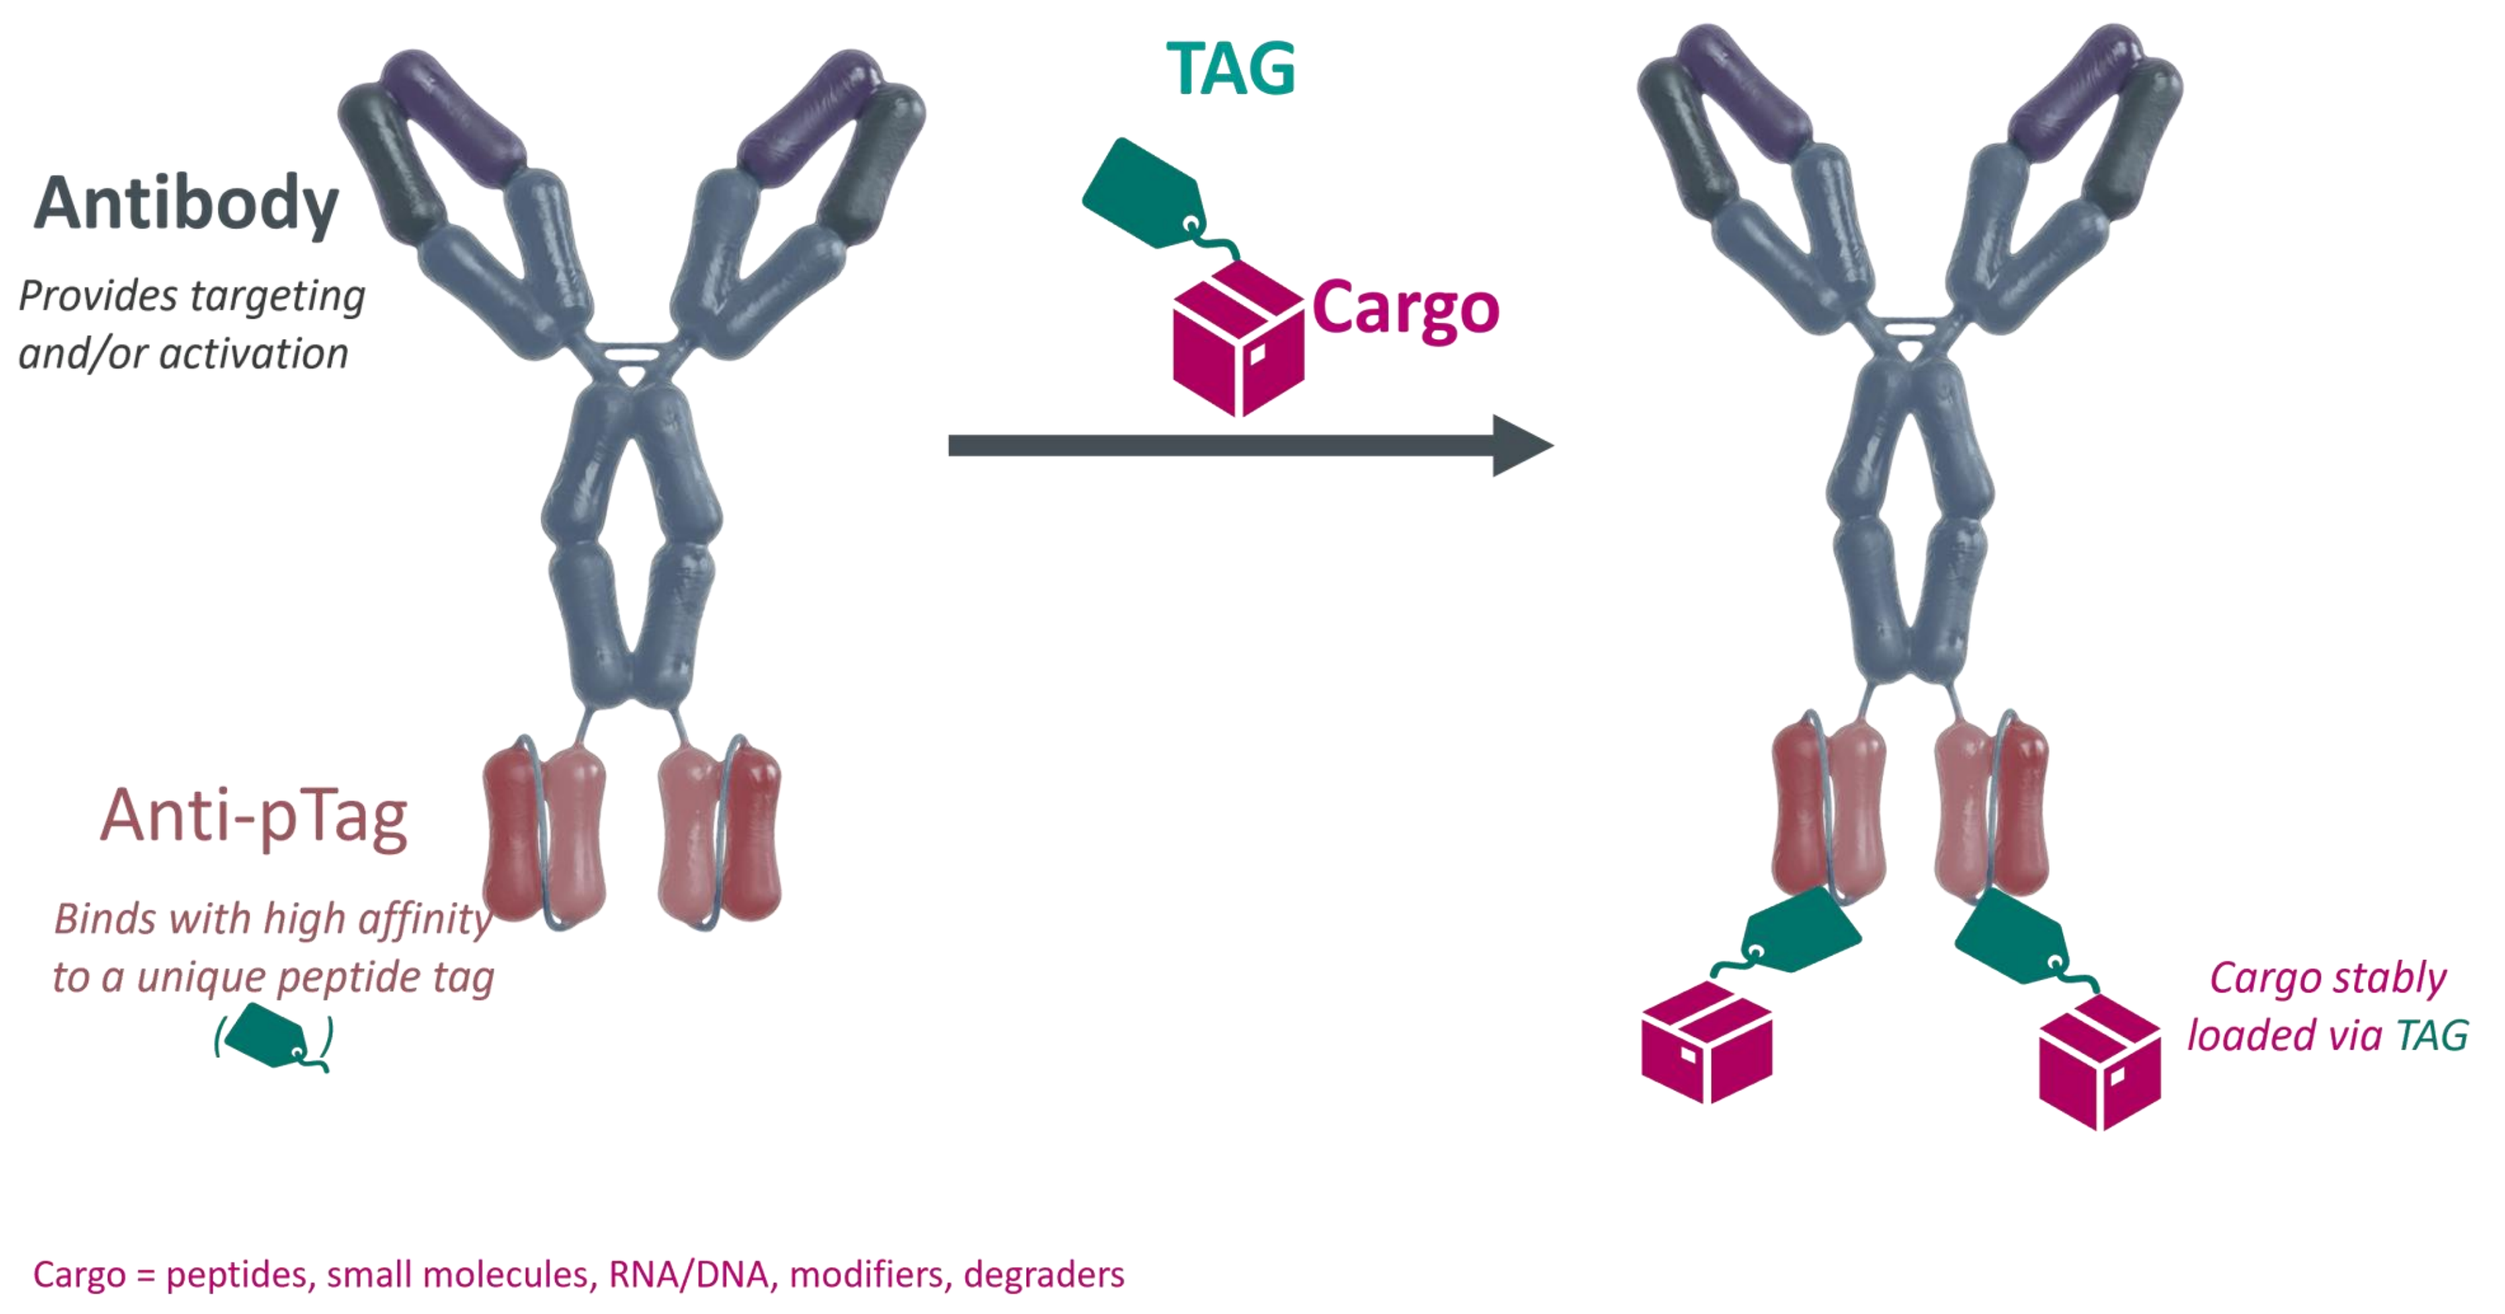
Antibody
Provides targeting and/or activation
TAG
Cargo
Anti-pTag
Binds with high affinity to a unique peptide tag
Cargo stably loaded via TAG
Cargo = peptides, small molecules, RNA/DNA, modifiers, degraders

1 An ADC-based approach targeting CD40 improves curative rates in a colorectal cancer model

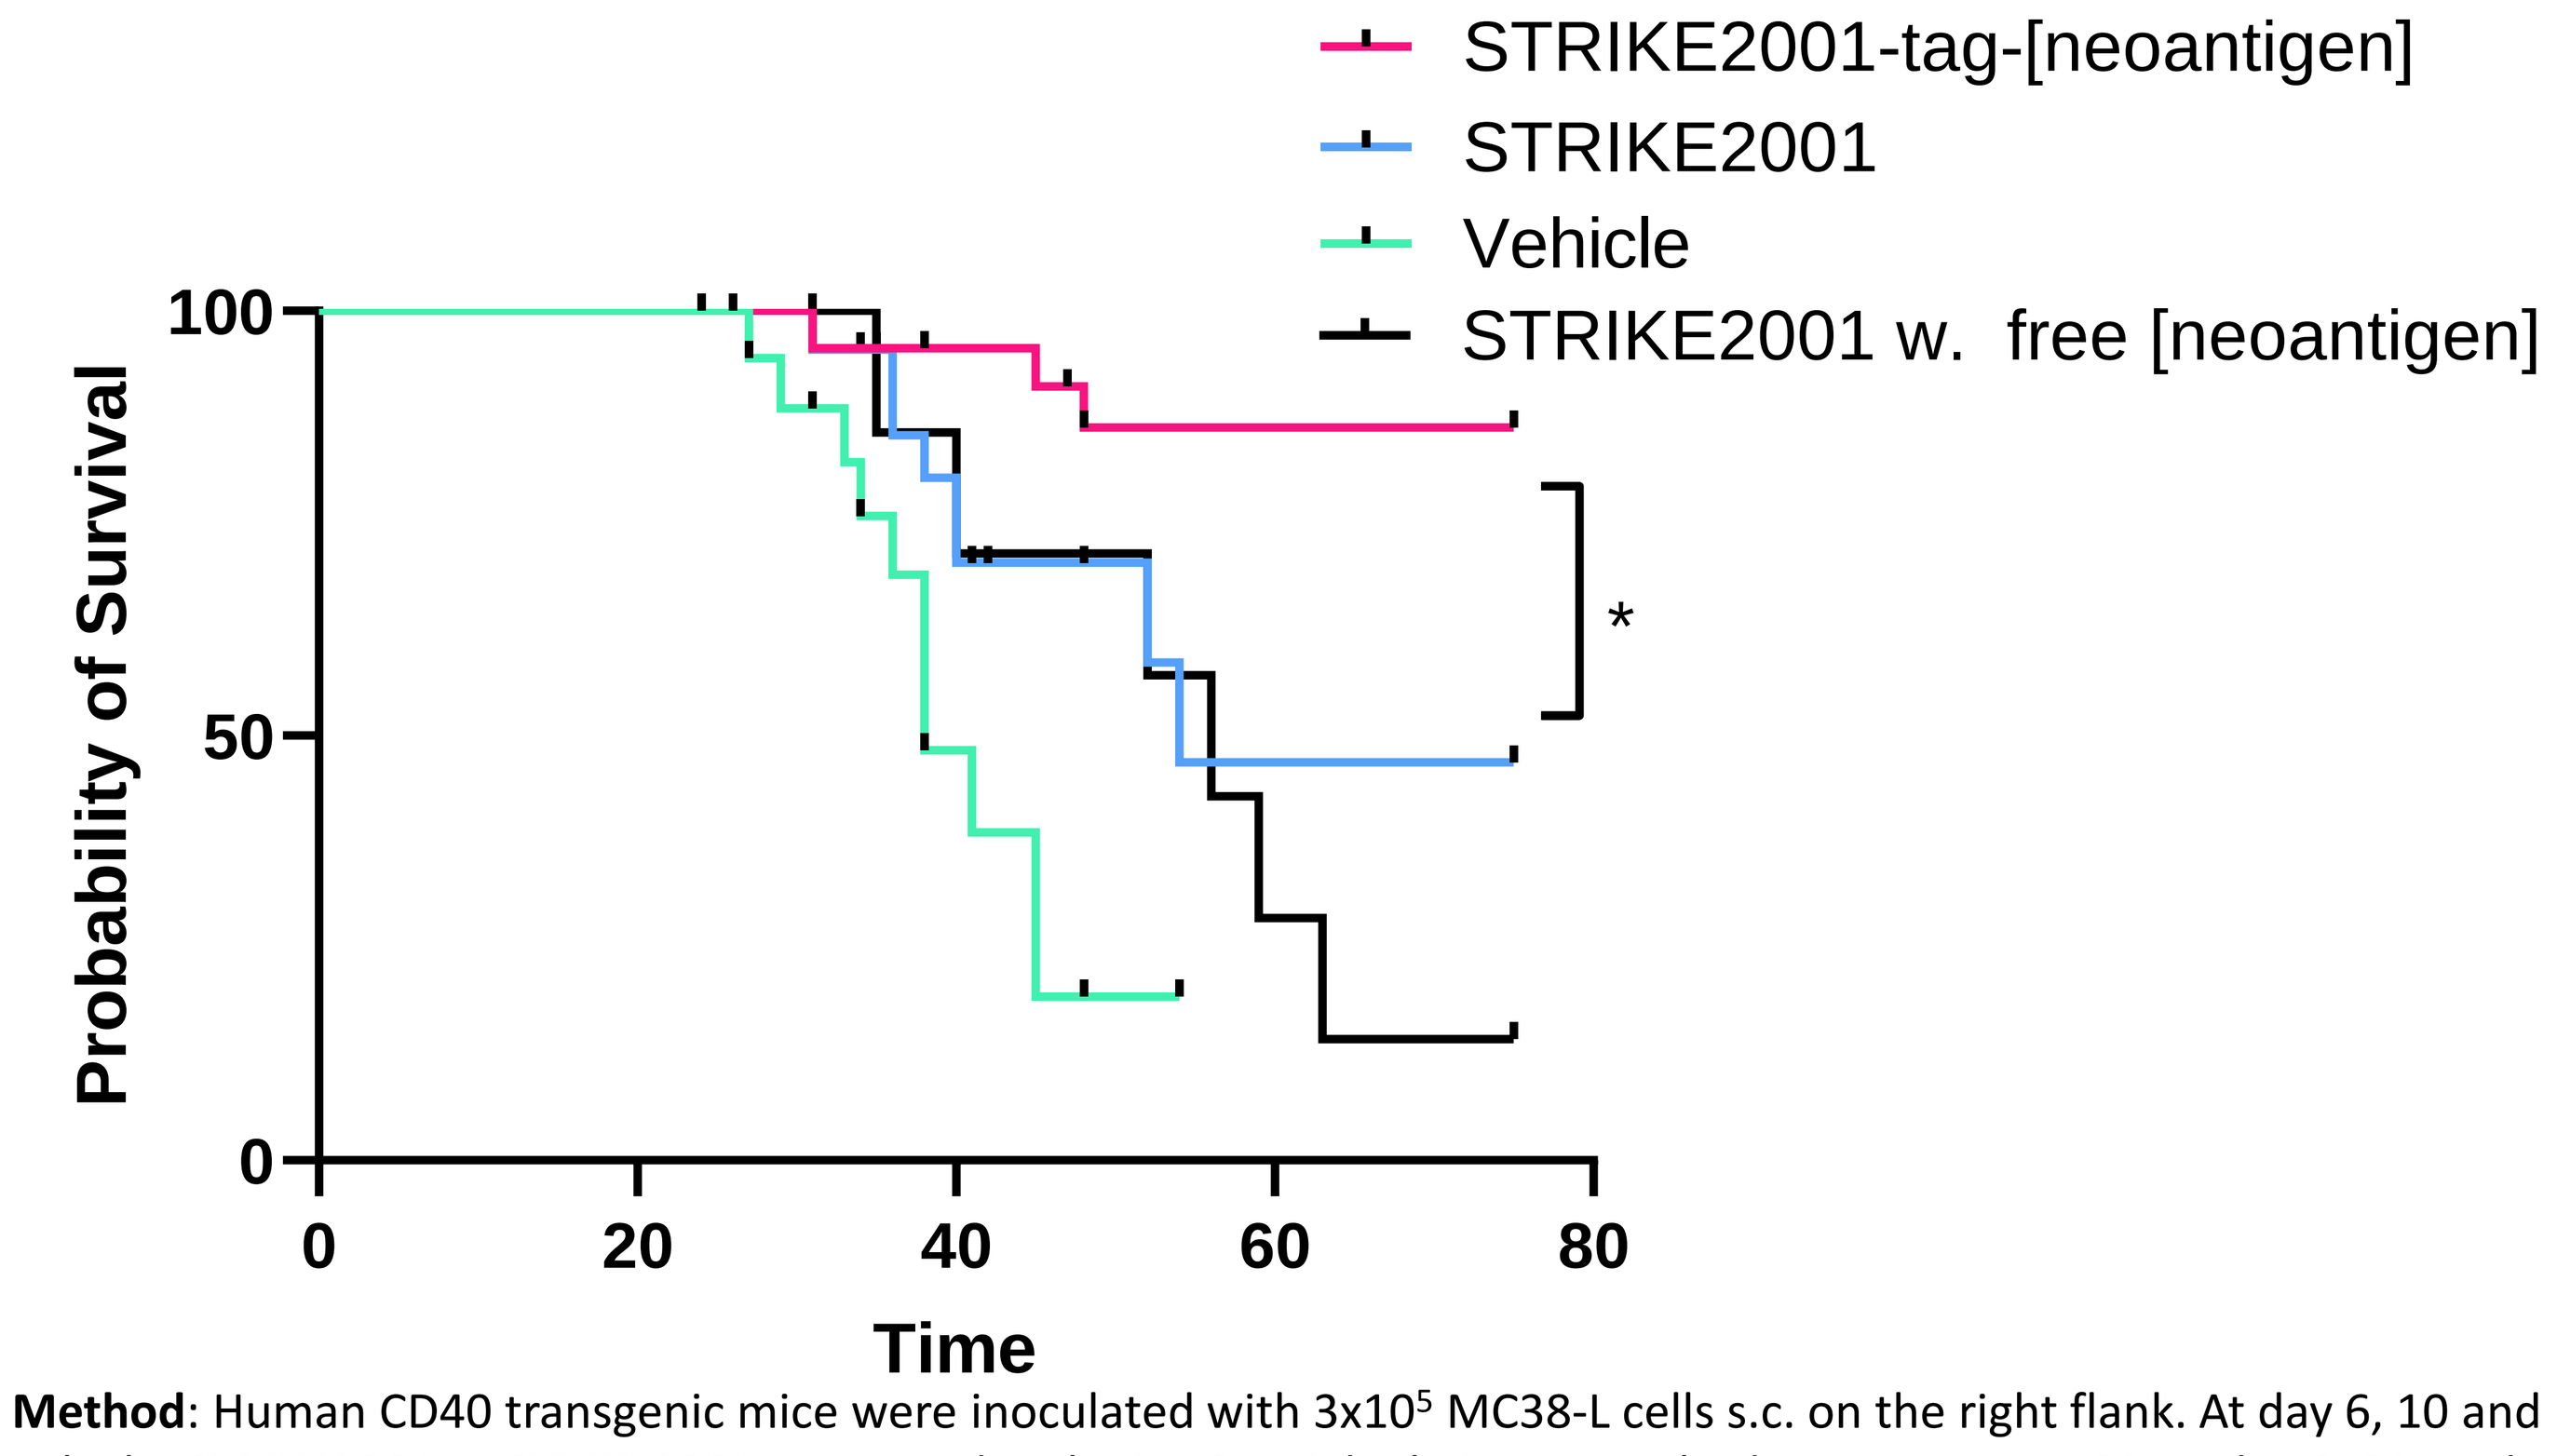
STRIKE2001-tag-[neoantigen]
STRIKE2001
Vehicle
STRIKE2001 w. free [neoantigen]
Probability of Survival
100
50
0
0
20
40
60
80
Time
*

Results: A previously identified neoantigen that has shown limited therapeutic efficacy when used as a sole neoantigen with alternative adjuvants in earlier published vaccination studies [1, 2] was used together with the CD40 agonistic antibody STRIKE2001 for targeted delivery of the neoantigen to CD40 expressing cells. Controls consisted of STRIKE2001 alone or loaded with an inert control antigen (blue line) or STRIKE2001 mixed with the neoantigen which could not bind STRIKE2001 (truncated tag, black line). The data represent two pooled experiments. **STRIKE2001 with linked neoantigen shows superior efficacy with a high rate of complete responders compared to STRIKE2001 alone or the non-linked antigen group.**

Method: Human CD40 transgenic mice were inoculated with 3x10^5 MC38-L cells s.c. on the right flank. At day 6, 10 and 14 post tumor inoculation, mice were treated s.c. at the left hock with either vehicle, STRIKE2001 or STRIKE2001 pre-mixed with pTag9aa-Adpgk. Doses antibody or conjugate: 30 µg day 6, 90 µg day 10 and 30 µg day 14. Tumor growth was monitored over time and mice were sacrificed either when they reached experimental endpoint (volume >1000 mm^3) or when they reached the humane endpoint (10% weight loss or ulcerated tumors, marked as censored in the graph). The survival curves between STRIKE2001 and STRIKE2001-tag-neoantigen differ, p<0.05 (Log rank test). Data is still maturing for one of the two experiments. 1. Hos, Brett J. et al. Cell Reports, Volume 41, Issue 2, 111485 2. Hos, B. J. et al (2019). OncoImmunology, 9(1).

2 Peptide tag variants show retained binding to STRIKE2001

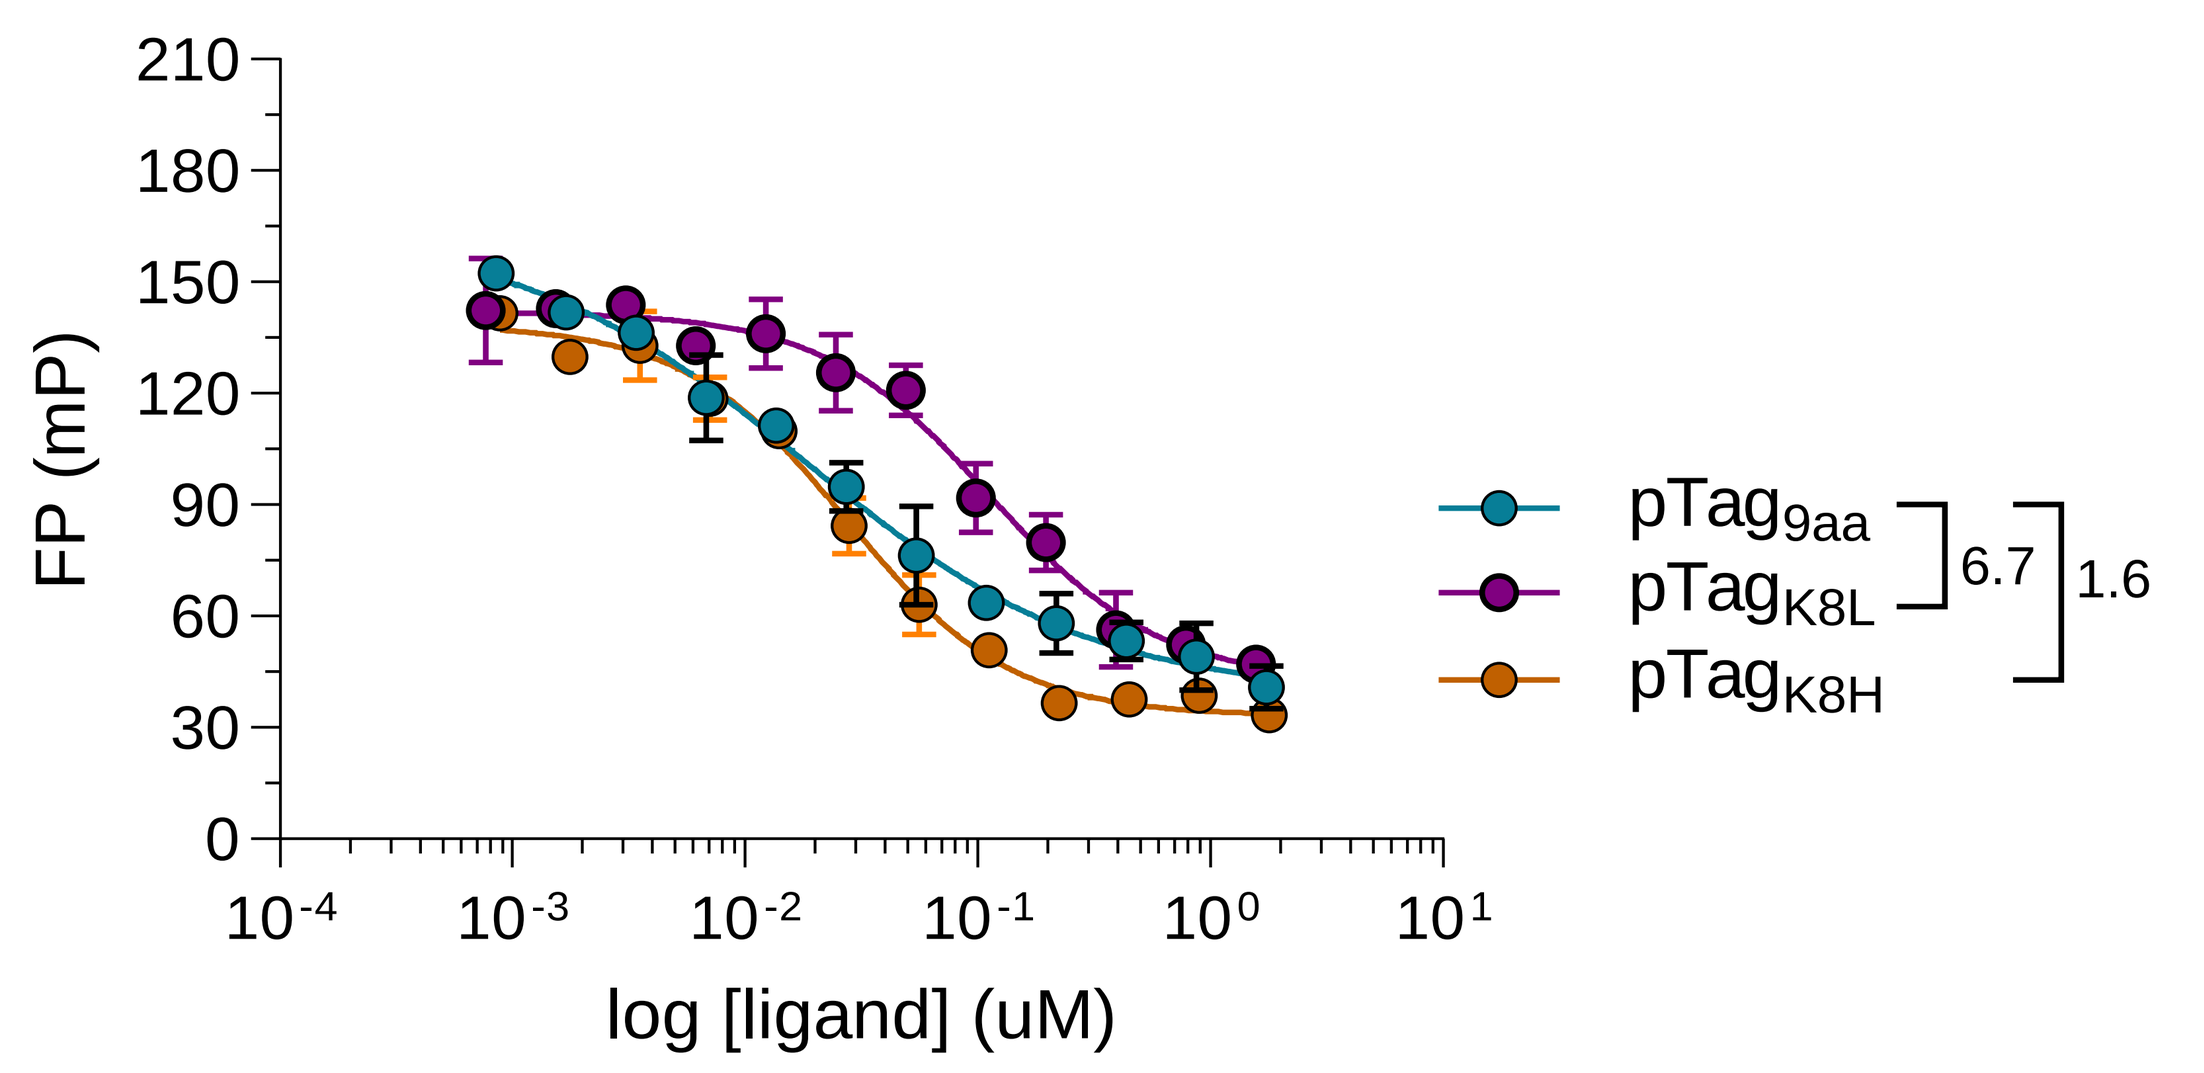
210
180
150
120
90
60
30
0
FP (mP)
10^{-4}
10^{-3}
10^{-2}
10^{-1}
10^{0}
10^{1}
log [ligand] (uM)
$pTag_{9aa}$
$pTag_{K8L}$
$pTag_{K8H}$
6.7
1.6

Results: Amino acid residues within an epitope that bind to an antibody are commonly essential to maintain high-affinity binding. The minimal epitope for retained binding to STRIKE2001 is an eight-amino-acid-long epitope. Here, we made use of the nine-amino-acid-long tag and altered residue eight to an amino acid with the same physicochemical properties (positively charged) as lysine (K), e.g., histidine (H), but also assessed an amino acid with a more aliphatic nature to further evaluate how this impacts biological responses to antigenic peptides. **STRIKE2001 binding to the tag is slightly impacted by the introduction of leucine (L), but affinities are still in the pM range.**

Method: The binding verification of various pTag variants. Binding and affinity of various pTags is measured by fluorescence polarization (FP), in which the displacement of the AF488-labeled pTag is analyzed. STRIKE2001 (3 nM) is premixed and saturated with the labeled pTag (2.5 nM). The unlabeled pTag-peptides of interest are added in an increasing concentration to the STRIKE2001 and AF488-labeled pTag. Over several days, the unlabeled peptide displaces the labeled peptide from STRIKE2001 and the FP signal will decrease. The numbers indicate the fold change in the calculated KD value. Estimated affinities: pTag9aa 9.4 pM, pTagK8L 63 pM and pTagK8H 15 pM.

3 The mutated tag carrying the aliphatic residue promotes CD8 T cell responses in the lower concentration range– *In vitro analyses*

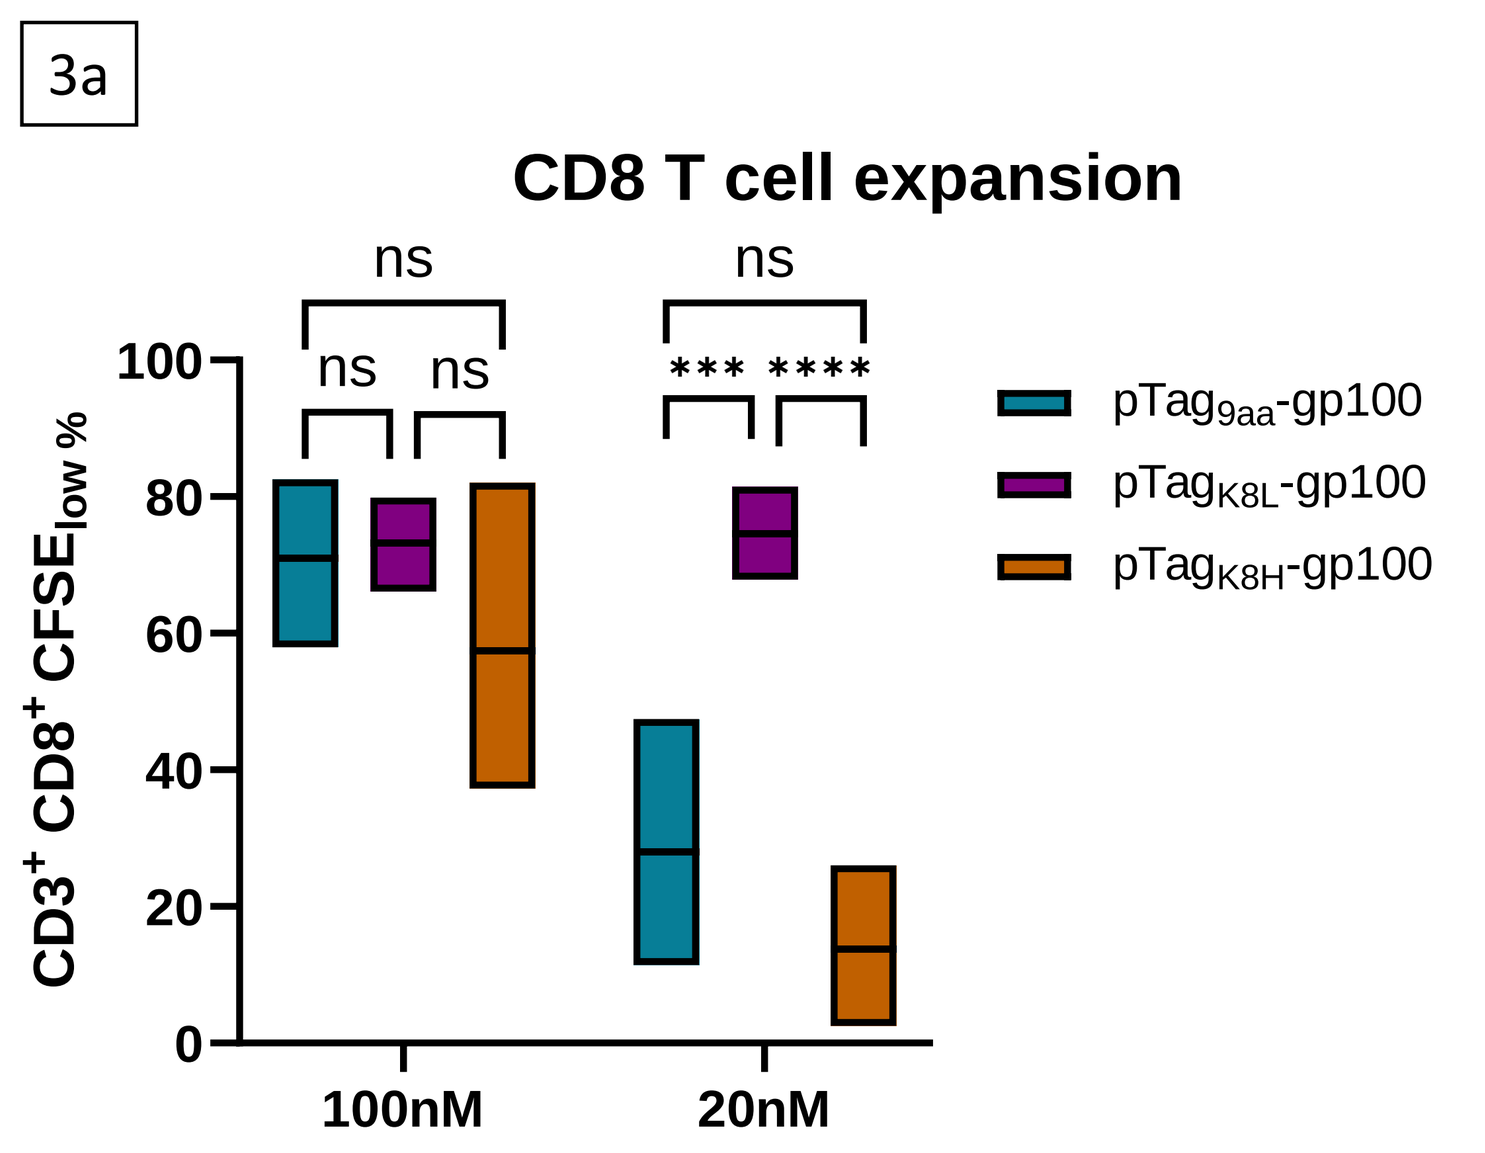
3a
CD8 T cell expansion
ns
ns
ns
ns
*** ****
100
80
60
40
20
0
$CD3^+ CD8^+ CFSE_{low}$ %
100nM
20nM
$pTag_{9aa}$-gp100
$pTag_{K8L}$-gp100
$pTag_{K8H}$-gp100

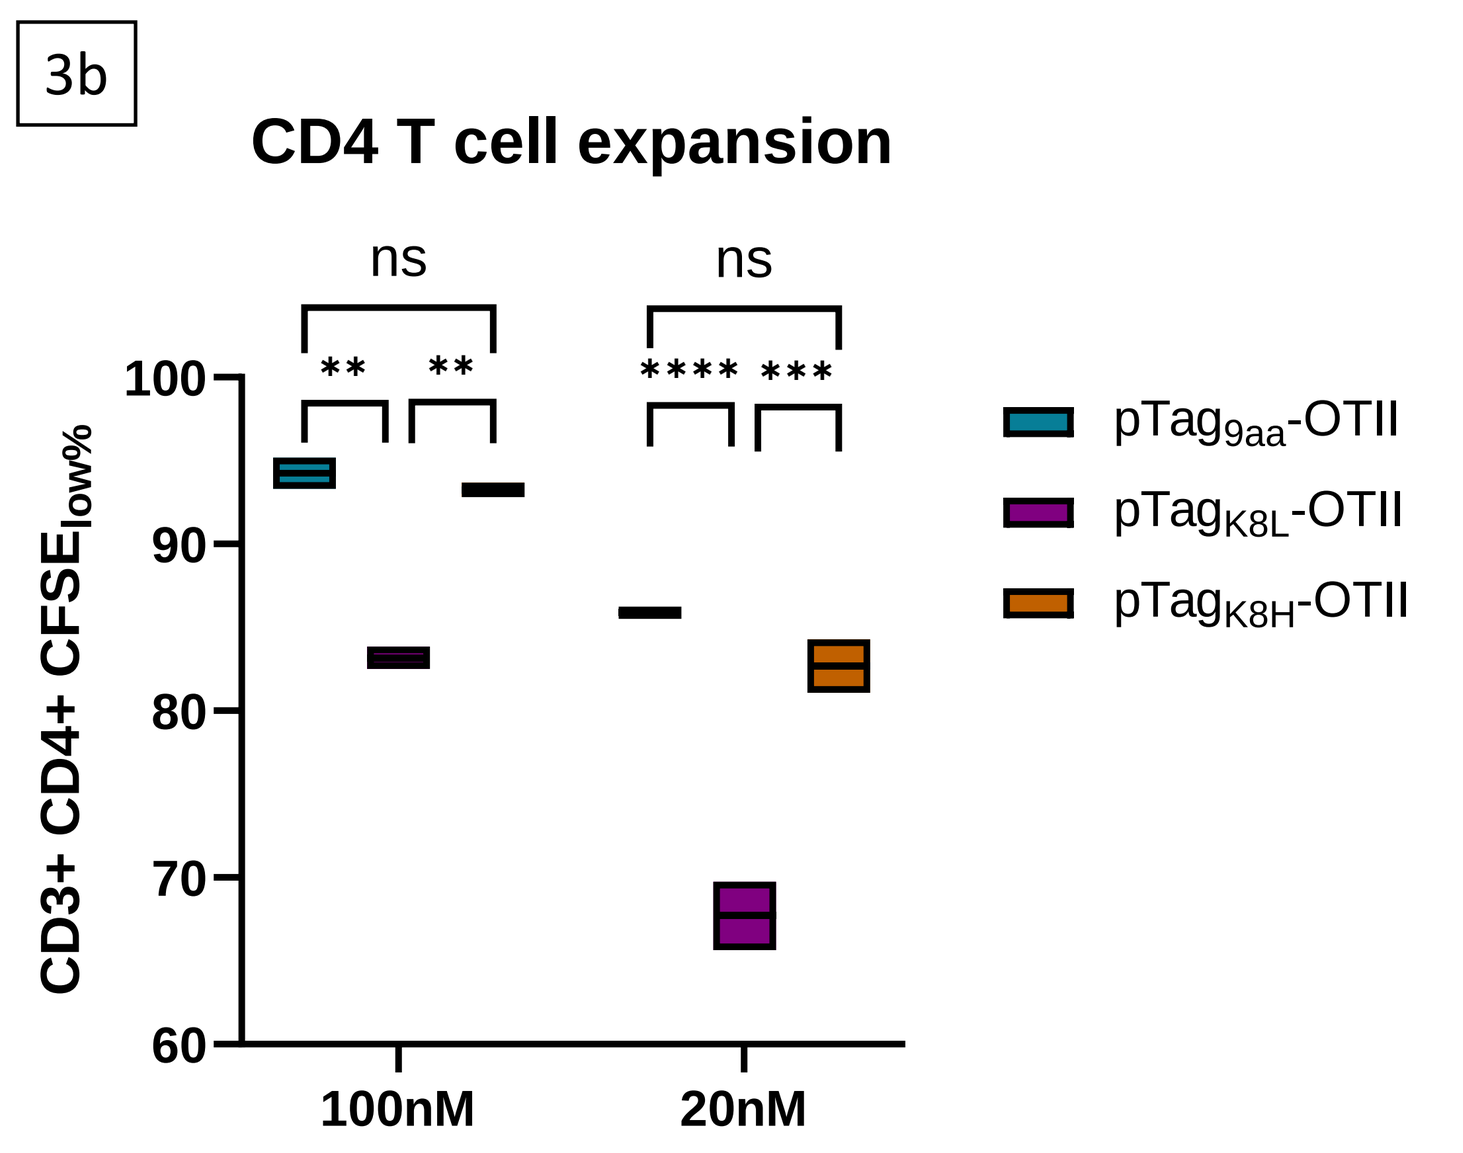
3b
CD4 T cell expansion
ns
ns
** **
**** ***
100
90
80
70
60
CD3+ CD4+ $CFSE_{low}$%
100nM
20nM
$pTag_{9aa}$-OTII
$pTag_{K8L}$-OTII
$pTag_{K8H}$-OTII

Results: Longer peptides carrying embedded immunogenic T cell epitopes are equally effective at expanding T cells in recall assay setups; e.g., the original tag does not impact antigen processing and presentation.

To evaluate if the tag variants impact antigen presentation, we studied T cell expansion in vitro using isolated CD8 or CD4 T cells that carry a TCR specific for the melanoma antigen gp100 (CD8 T cell responses) or the ovalbumin antigen (CD4 T cell responses).

The pTagK8H did not alter T cell activation patterns compared to the original pTag sequence; however, the **pTagK8L with the aliphatic amino acid residue change appears to improve CD8 responses in the lower concentration range and negatively impact CD4 responses at both concentrations tested.** As CD8 responses depend on cross-presentation (i.e., the antigen leaves the endosome to enter the cytoplasm), while CD4 responses depend on loading of the epitope to MHC class II within the endosome, it is plausible that the residue change impacts endosomal escape. Alternative explanations are presented in the figure to the right.

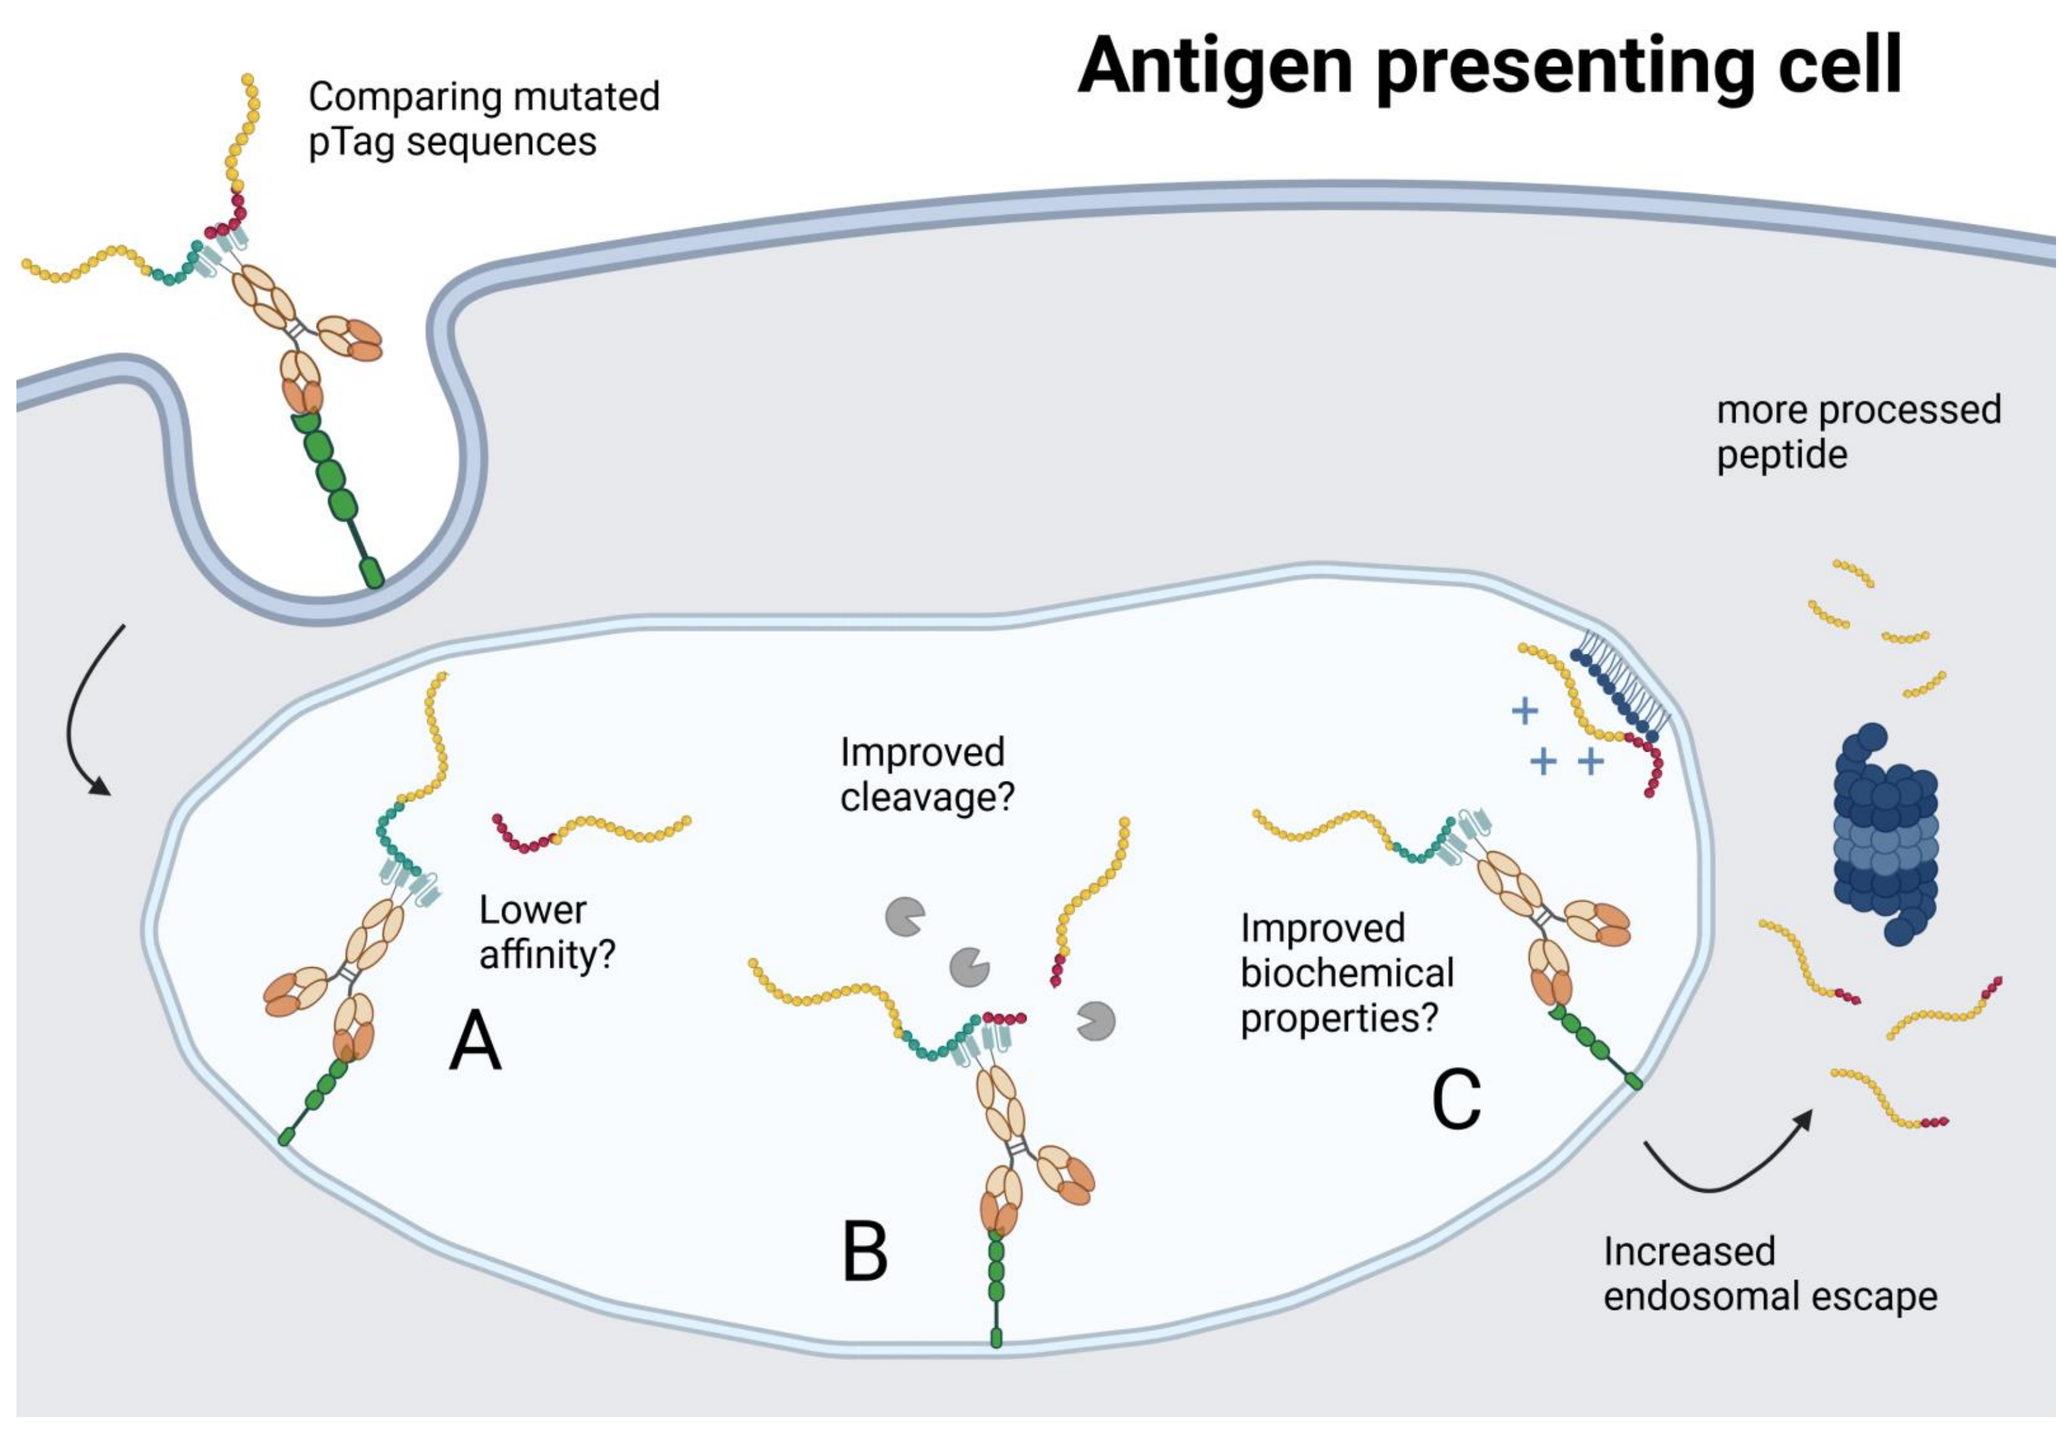
Antigen presenting cell
Comparing mutated pTag sequences
more processed peptide
Improved cleavage?
Lower affinity?
Improved biochemical properties?
A
B
C
Increased endosomal escape

Method: Cultured bone marrow derived dendritic cells (BMDCs) of transgenic human CD40 mice were incubated for 24 hours with either 100 nM or 20 nM of the different pTags conjugated to either the gp100 peptide (CD8 epitope, Figure 3a) or OTII peptide (CD4 epitope, Figure 3b). After 24 hours incubation, the BMDCs were washed to remove any remaining peptides. Either CD8 T cells isolated from the spleen of transgenic PMEL mice (T cell receptor specific for gp100 peptide) or CD4 T cells isolated from the spleen of transgenic OTII mice (T cell receptor specific for OTII peptide) were stained with CFSE and added to the peptide-loaded BMDCs at a 1:1 ratio. After 72 hours of co-culture, cells were harvested analyzed by flow cytometry. High proliferating CD8 and CD4 T cells "dilute out" the CFSE staining and were therefore characterized as CD3+ CD8+ $CFSE_{low}$ cells and CD3+ CD4+ $CFSE_{low}$ cells. Data of both CD8 and CD4 T cell expansion represents two replicate experiments. Statistical analysis was performed by using a 2-way ANOVA with Tukey's multiple comparisons test.

4 CD4 but not CD8 responses differ between the tags when studying T cell responses *in vivo*

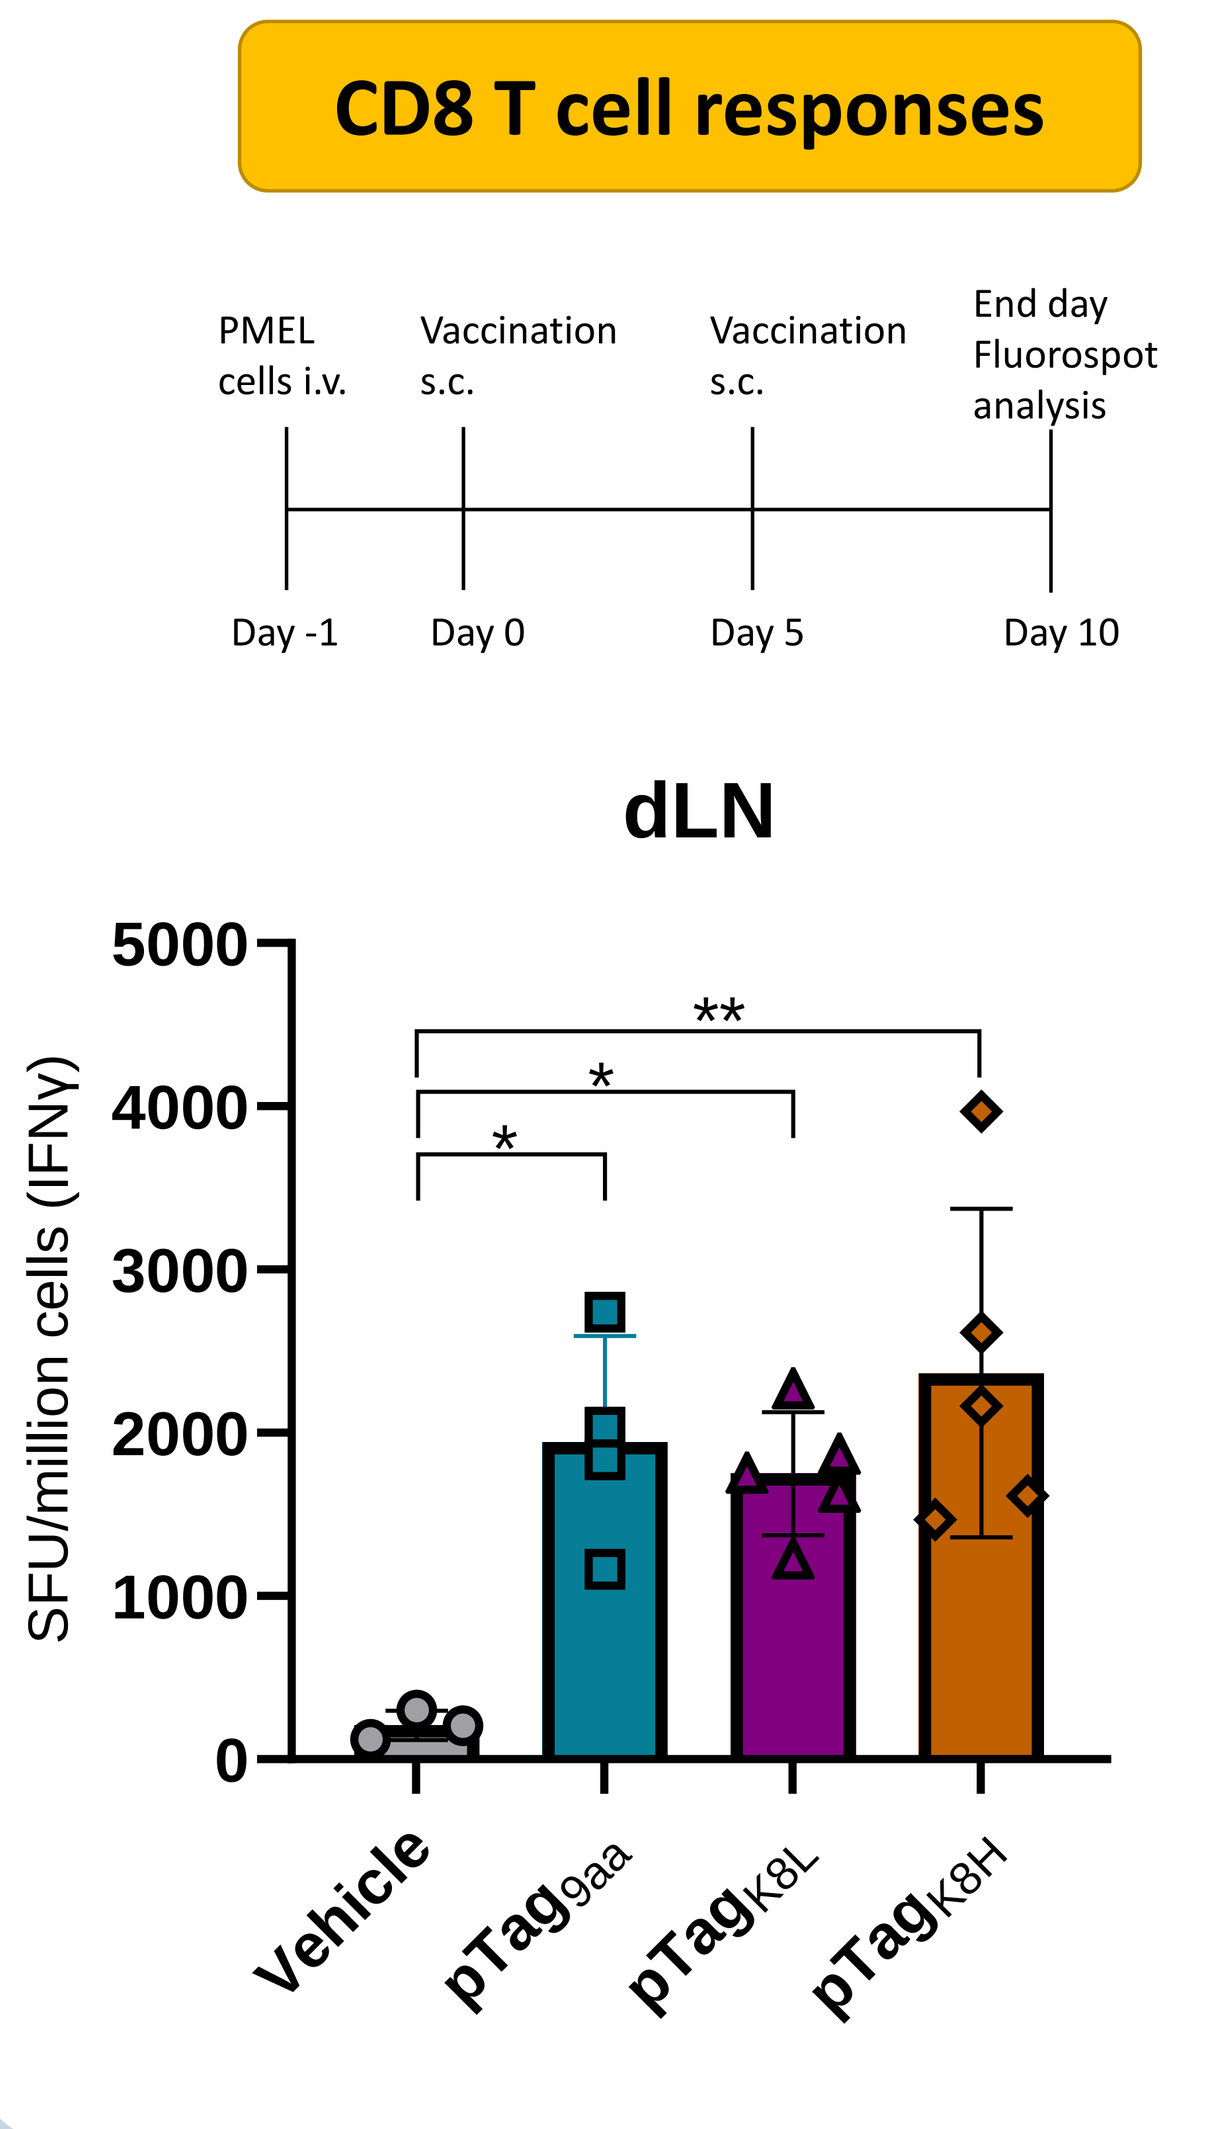
CD8 T cell responses
PMEL cells i.v.
Vaccination s.c.
Vaccination s.c.
End day Fluorospot analysis
Day -1
Day 0
Day 5
Day 10
dLN
5000
4000
3000
2000
1000
0
SFU/million cells (IFNγ)
**
*
*
Vehicle
$pTag_{9aa}$
$pTag_{K8L}$
$pTag_{K8H}$

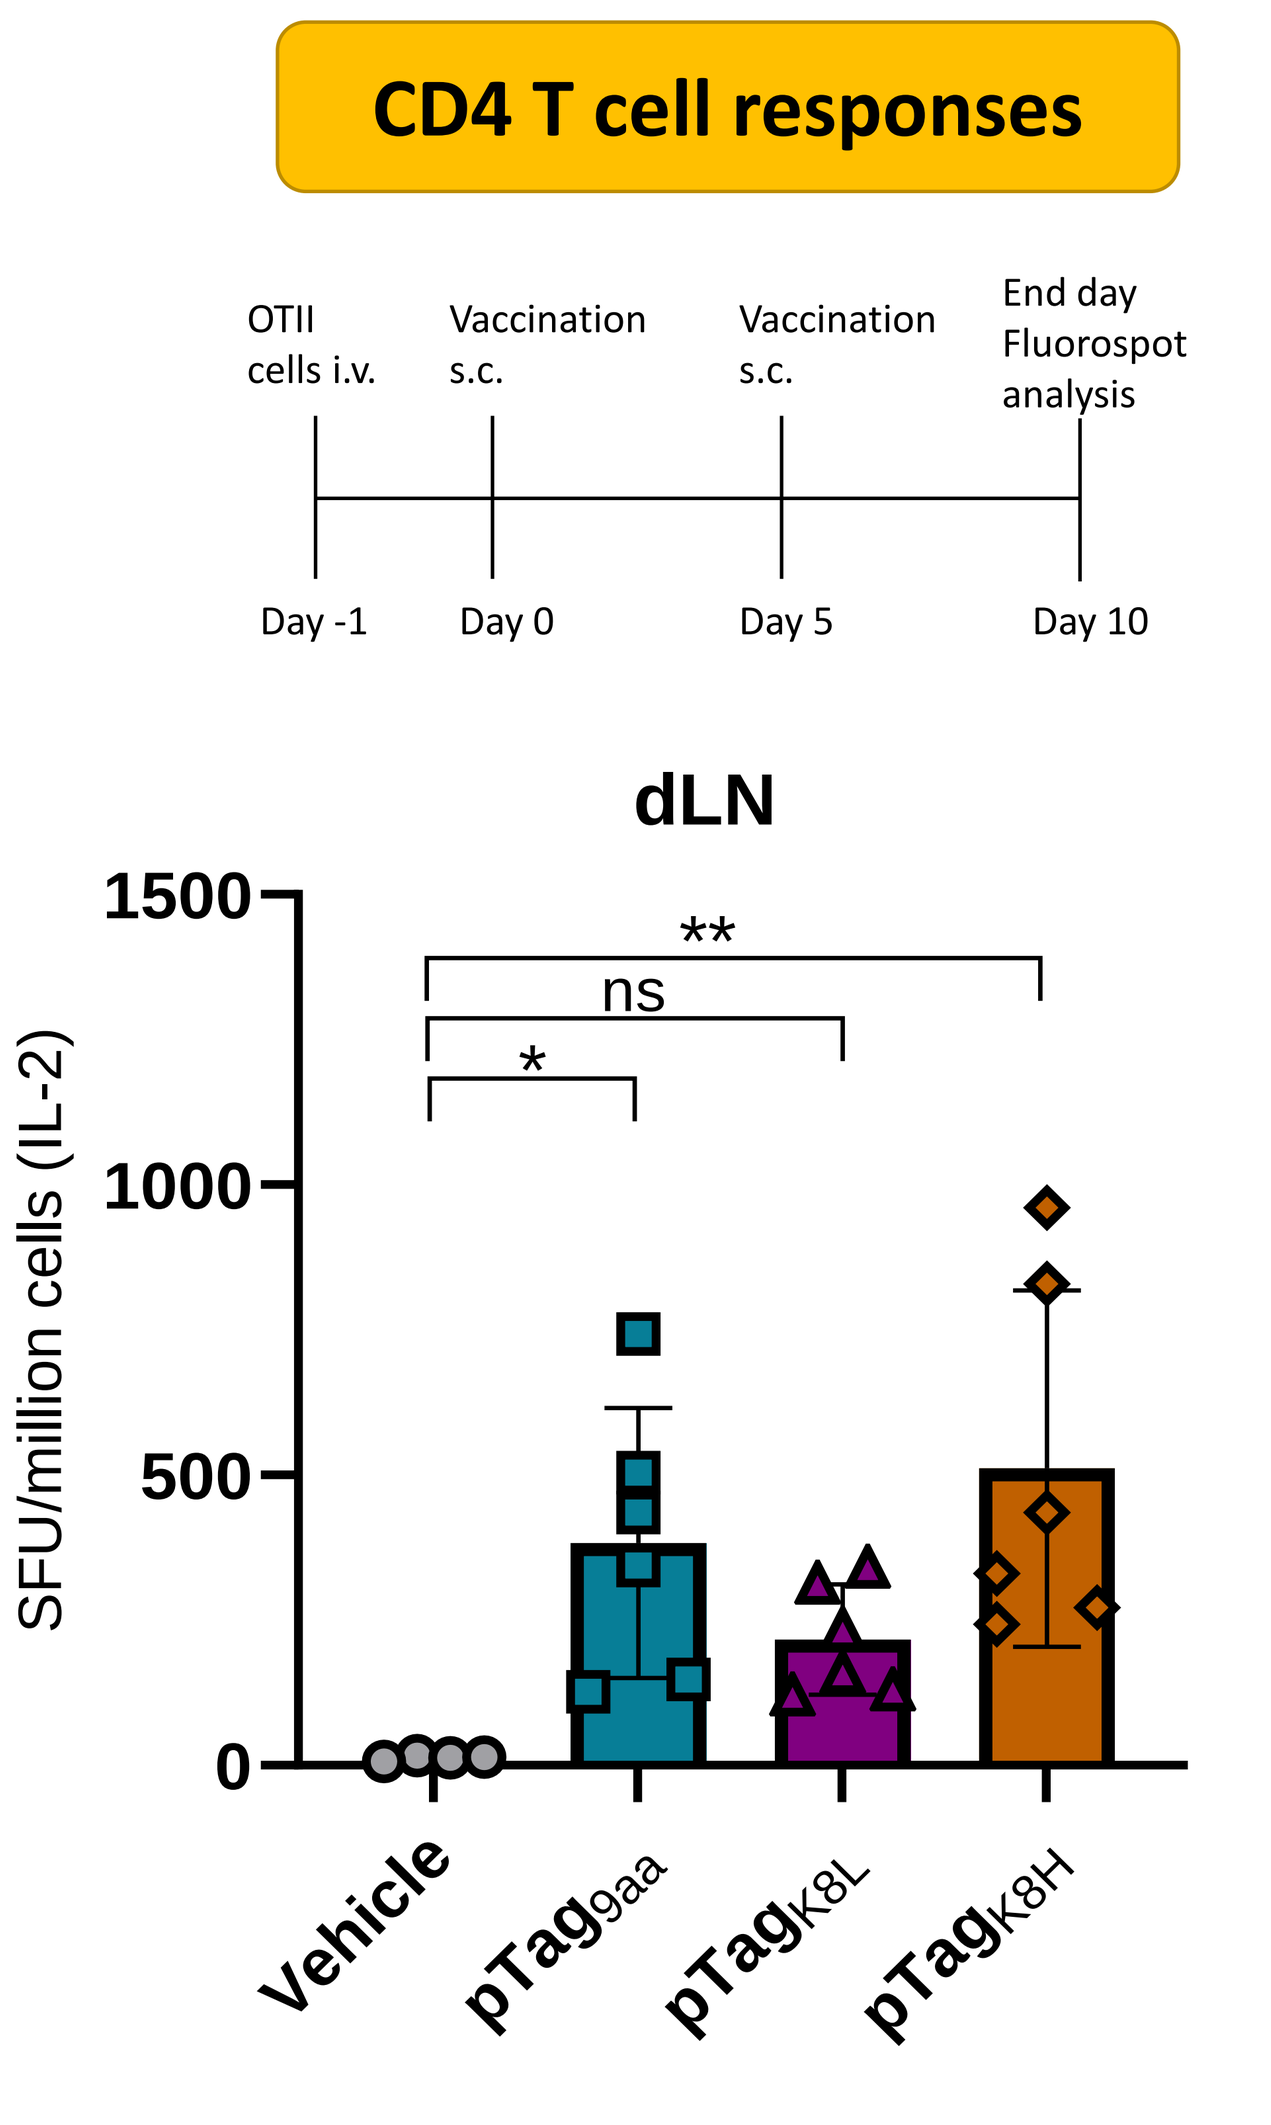
CD4 T cell responses
OTII cells i.v.
Vaccination s.c.
Vaccination s.c.
End day Fluorospot analysis
Day -1
Day 0
Day 5
Day 10
dLN
1500
1000
500
0
SFU/million cells (IL-2)
**
ns
*
Vehicle
$pTag_{9aa}$
$pTag_{K8L}$
$pTag_{K8H}$

Results: To verify the in vitro results, we performed an in vivo experiment using isolated antigen-specific T cells for gp100 or ovalbumin (as used in the in vitro experiment above). The assay was performed with the tagged peptides formulated with STRIKE2001.

CD8 T cell activation did not differ between the tagged peptide versions, while the CD4 responses mimicked the in vitro results, with the aliphatic residue negatively impacting CD4 responses. We hypothesized that the delivery via STRIKE2001 impacted the results, as the agonistic activity of the antibody also promotes cross-presentation. As such, we performed another in vitro experiment without STRIKE2001, using tagged but free peptide combined with adjuvants (LPS or CpG ODN). LPS was too weak of an adjuvant to induce T cell responses, and with CpG stimulation, T cells were expanded but without a noted difference between the tag variants. It is thus plausible that the impact the tag variant has on antigen presentation is only relevant in suboptimal concentration ranges. We plan to follow up with an experiment to evaluate responses to a single administration (prime dose only), as all previous in vivo experiments were performed using the prime-boost schedule.

Method: hCD40 transgenic mice were i.v. injected with 100x10^6 splenocytes derived from either transgenic PMEL mice (CD8 T cells specific for gp100$^{25\text{-}33}$ peptide) or transgenic OTII mice (CD4 T cells specific for OVA$^{329\text{-}337}$ peptide). hCD40 mice were vaccinated twice s.c. hock with pre-mixed STRIKE2001 (30 µg/150 pmol) and the different pTags conjugated to the gp100$^{25\text{-}33}$ peptide (1.46 µg/450 pmol) or the OVA$^{329\text{-}337}$ peptide (1.88 µg/450 pmol). On day 10, mice were sacrificed and lymph nodes at the draining site (dLN) and spleen were collected. Next, the dLN & spleen mixed cells were added to either an IFNγ fluorospot (CD8) or an IL-2 fluorospot (CD4) and the cells were restimulated with either the gp100$^{25\text{-}33}$ or OVA$^{329\text{-}337}$ peptide (5 µM). After 20-hour, the fluorospot plates were developed. Data is plotted as spot forming units (SFU) per million cells. Data of both CD8 and CD4 T cell expansion represents one experiment with 4-6 mice per group. Statistical analysis was performed by using a one-way ANOVA with Holm-Sidak's multiple comparisons test.

Conclusion

ADCs can promote targeted delivery of drugs, thereby improving efficacy and reducing toxicity. For drug entities with a physicochemical nature that makes them prone to degradation (such as peptides and unmodified oligonucleotides) or that are highly negatively charged, leading to class-based exposure to certain organs while not reaching other sites, it is essential to develop new methods for drug delivery and intracellular routing.

We have developed an ADC-like method that can be adapted with various cargos, such as peptide-based neoantigens, and we have demonstrated superior anti-tumor efficacy compared to antibody alone using this technology. **Here, we show that amino acid residues in the tag element can be altered without significant impact of the binding to the antibody. The introduction of an aliphatic residue appears to promote CD8 T cell responses and reduce CD4 T cell responses**, indicating improved cross-presentation properties of the antigenic cargo, possibly relevant in situations with limited drug availability. Future studies will investigate if additional amino acid sequences outside the tag epitope, as linker elements or spacers, can further promote endosomal escape/cross-presentation properties in vivo when combined with the targeted delivery strategy presented herein.